Interdisciplinary Collaboration Training for BCBAs and Special Education Teachers to Support Young Children with Autism

Crystal Orta, Heidy Garcia, Jennifer Symon, Ya-Chih Chang
California State University, Los Angeles

INTRODUCTION

Early childhood special education (ECSE) teachers and related service providers experience challenges in collaboration due to limited training in their pre-service programs in collaborative partnerships.

The federally funded PISCES program was developed at California State University, Los Angeles, a diverse, urban institution that serves a predominantly traditionally underrepresented student population.

The goal of the program was to improve collaborative teaming between ECSE teachers earning a MA in Special Education and behavior analysts earning an MS in Counseling, in Applied Behavior Analysis along with a university certificate in Autism Spectrum Disorders (ASD).

"PISCES" PROGRAM

Partners in Improving Social Communication in Early Childhood Special Education program (PISCES) is an interdisciplinary preservice training program in Applied Behavior Analysis and Early Childhood Special Education.

Scholars participate in their regular courses of study in their respective programs. They also complete 4 shared courses: Assessment in special Education, Advanced Strategies in ASD, Augmentative and Alternative Communication and Fieldwork in ASD. Fieldwork focused on understanding key features of ASD and developmental milestones, creating developmentally appropriate goals, using evidence-based interventions, monitoring students' progress, and developing socially meaningful outcomes.

Training content and teaching opportunities for scholars involves learning evidence-based interventions to increase social communication in young children with ASD and significant communication delays.

Participants also attend bi-annual seminars with interdisciplinary experts, families, educators and related service providers.

CURRENT STUDY

The current study:
(1) Presents the interdisciplinary training program for ECSE and ABA scholars in the PISCES program.
(2) Presents on the identified 7 themes that emerged throughout the fieldwork sessions.
(3) Presents the frequency of themes meaningfully discussed during group supervision seminar sessions.

PARTICIPANTS

Fourteen participants were included in the study. All participants are graduate students from a diverse, minority-serving institution located in Los Angeles. Participants are enrolled in the master's program in Special Education, option in Early Childhood Special Education (n=5), or the master's program in Counseling, option in Applied Behavior Analysis (n=9). Participants are of diverse backgrounds (64% Hispanic, 21% White, 14% Asian). Eighty-six percent of the participants are female. All participants also work part-time or full-time with young children with autism spectrum disorders (ages 2-11) in various settings including home and school .

Ethnicity	Hispanic 64% White 21% Asian 14%
Gender	Female 86% Male 14%
Role	Behavior analyst 57% University Intern 21% Student teacher 14% ABA supervisor 7%
Setting	Home 50% School 43% Daycare 7%

METHODS

The study analyzed previous data collected by Chang, Garcia, Orta, Symon and Fryling (2022). The data included data collected from five online group seminars (60 min) that took place throughout a 15-week semester. The sessions were video recorded and transcribed. Subsequently, the transcriptions of the participant's beneficial topic contributions were reviewed and coded. Researchers identified main themes that emerged from the interactions between scholars and university faculty. Seven frequent themes emerged and are presented. The thematic frequency of each of the 5 sessions were coded and calculated.

The study was funded by OSEP #H325K180154
and Cal State LA College of Education

RESULTS: THEMES THAT EMERGED

The following themes emerged from the collaborative fieldwork seminars

1. The importance of writing and targeting socially meaningful and developmentally appropriate goals
2. Developing goals that relate to the DSM5 criteria of ASD
3. Using various assessments and collecting data
4. Asking and sharing input/feedback from different disciplines (ECSE and ABA)
5. Consideration of cultural of and family implication
6. Knowledge of ASD and Training
7. Evidence Based Practice in ASD (strategies/interventions)

RESULTS: FREQUENCY OF THEMES

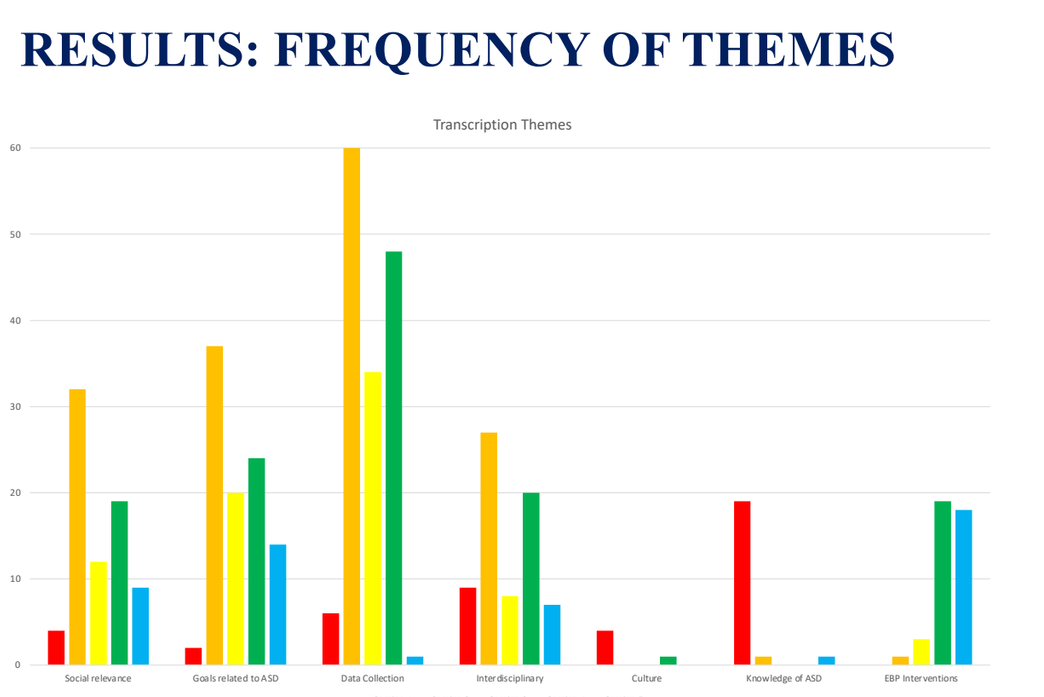
Transcription Themes

IMPLICATIONS/FUTURE RESEARCH

- A strong emphasis was placed on goal development, including socially meaningful goals that target core features of ASD
- Interdisciplinary collaboration was a theme present across each group supervision session.
- Results show limited discussion related to cultural diversity and family responsiveness. In the future, this theme will be specifically added into fieldwork assignments.
- Future studies should examine long-term impact of these collaborative programs, particularly within scholars' professional settings.

Enhancing Care to Families Affected by Autism Through Compassion, Culture, and Trauma-Informed Behavioral Approaches

E. Azadeh Hanukaai, Martin Valdez, Silfia Chatibus Satari, Debra Berry Malmberg

Introduction

- Board Certified Behavior Analysts (BCBAs) must abide to the Behavior Analyst Certification Board (BACB) Code of Ethics (2020). The core principles state to maximize benefits, do no harm, and to treat others with compassion, dignity and respect. Ethical BCBAs must incorporate these principles in everyday practice to provide participants with most effective treatment.
- Compassionate care skills demonstrate high social validity correlations with patient satisfaction, treatment adherence, and improved clinical outcomes in both medicine and mental health (Hojat et al., 2011).
- Highly rated physicians on the Jefferson Scale of Empathy were correlated with greater clinical outcomes in regard to behavior management of diabetes (Hojat et al., 2011)
- Behavior analysts with poor relationship skills result to family dissatisfaction. This is linked to ethical complaints filed by parents due to lack of interpersonal skills (Taylor et al, 2019)
- Trauma Informed Care (TIC) interventions have shown improvements on emotional and behavioral wellbeing, while reducing challenging behaviors with children in the welfare system (Zhang et al., 2021)
- Enhancing care through trauma informed care, culture, and compassion will lead to higher well being with the clients we serve.

- **Purpose**: Conduct a literature review to teach compassionate, cultural, and trauma-informed care and its application with clients and families, summarizing recommendations for the field of behavior analysis to abide by the BACB ethics code to provide the most effective treatment while doing no harm.

Operational Definitions

- Trauma results from an individual's experiences with certain events or circumstances that were physically or emotionally harmful or life threatening and that can have lasting aversive impact on the individual's well-being. (SAMHSA, 2014)
- Trauma Informed Care are informed approaches to therapy that focus on an individual's specific lived circumstances promoting a culture of safety, healing and overall well being. Approaches acknowledge trauma's impact and promotes control, choice, and autonomy. (SAMHSA, 2014)
- Culturally Responsive Care are informed approaches to therapy that focus on developing an intentional respect of a person's social, cultural and linguistic needs to provide socially meaningful services. (Munoz, 2007, Jimenez-Gomez & Beaulieu, 2022, Bentancourt et al, 2002)
- Compassionate Care Skills are informed approaches to therapy that focus on interpersonal skills and strategies for building relationships with families. Skills focus on understanding the individual's situation, perspective, and feelings, communicating with empathetic understanding, and checking for accurate understanding (Taylor et. al, 2019)

Figure 1

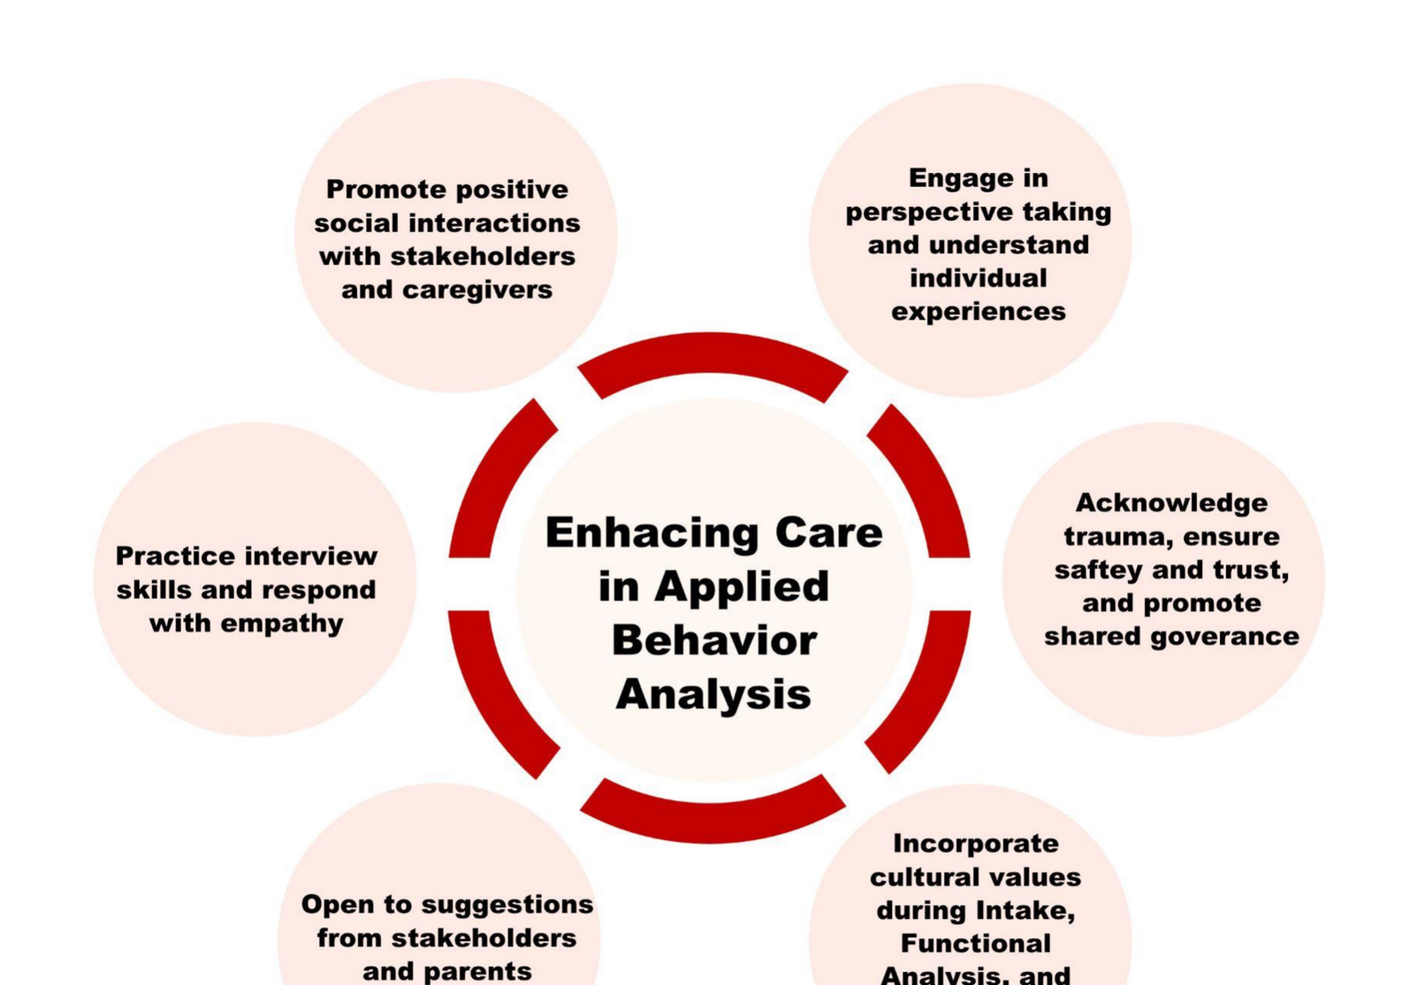
Enhacing Care in Applied Behavior Analysis
Promote positive social interactions with stakeholders and caregivers
Engage in perspective taking and understand individual experiences
Acknowledge trauma, ensure saftey and trust, and promote shared goverance
Incorporate cultural values during Intake, Functional Analysis, and Intervention process.
Open to suggestions from stakeholders and parents
Practice interview skills and respond with empathy

Methods

- Search Google Scholar with keywords, "Trauma", "Trauma Informed Care", "Cultural Competence", "Culture", "Compassionate Care," "Compassionate Care Skills", "Applied Behavior Analysis", and "ABA".
- Inclusionary Criteria: Any empirical article, review article or book chapter published in English that included keywords.
- Review the results of studies and/or recommendations provided to inform clinical recommendations.

References

Behavior Analyst Certification Board. (2020). *Ethics code for behavior analysts*. Littleton, CO: Author.
Betancourt, Joseph R., et al. *Cultural Competence in Health Care: Emerging Frameworks and Practical Approaches*. Commonwealth Fund, Quality of Care for Underserved Populations, 2002, pp.3.
Hojat, Mohammadreza, et al. "Physicians' Empathy and Clinical Outcomes for Diabetic Patients." *Academic Medicine*, vol. 86, no. 3, 2011, pp. 359–364., https://doi.org/10.1097/acm.0b013e3182086fe1.
Jimenez-Gomez, Corina, and Lauren Beaulieu. "Cultural Responsiveness in Applied Behavior Analysis: Research and Practice." *Journal of Applied Behavior Analysis*, pp. 650–673., https://doi.org/10.1002/jaba.920.
Muñoz, Jaime Phillip. "Culturally Responsive Caring in Occupational Therapy." *Occupational Therapy International*, pp. 256–280., .
https://doi.org/10.1002/oti.238.
Rajaraman, Adithyan, et al. "Toward Trauma-Informed Applications of Behavior Analysis." *Journal of Applied Behavior Analysis*, 1, 2021, pp. 40–61., https://doi.org/10.1002/jaba.881.
Rohrer, Jessica L., and Mary Jane Weiss. "Teaching Compassion Skills to Students of Behavior Analysis: A Preliminary Investigation." *Behavior Analysis in Practice*, 2022, https://doi.org/10.1007/s40617-022-00748-y.
Taylor, Bridget A., et al. "Compassionate Care in Behavior Analytic Treatment: Can Outcomes Be Enhanced by Attending to Relationships with Caregivers?" *Behavior Analysis in Practice*, 2018, pp. 654–666., https://doi.org/10.1007/s40617-018-00289-3.
Substance Abuse and Mental Health Services Administration. SAMHSA's Concept of Trauma and Guidance for a Trauma-Informed Approach. HHS Publication No. (SMA) 14-4884. Rockville, MD: Substance Abuse and Mental Health Services Administration, 2014.
Zhang, Saijun, et al. "Trauma-Informed Care for Children Involved with the Child Welfare System: A Meta-Analysis." *Child Abuse & Neglect*, p. 105296., https://doi.org/10.1016/j.chiabu.2021.105296.

Clinical Recommendations

Compassionate Care Skills (2.15, 3.01)

- Basic interviewing skills: positive introduction, note-taking, back channeling, communicative efforts demonstrating active listening
- Interest in family: acknowledge client's abilities and interests, ask stakeholder priorities and preferences to incorporate into intervention.
- Collaborate with family: normalize parent experiences and collaborate with stakeholders to promote client success. (Rohrer & Weiss, 2022)
- Promote positive social interactions: maintain eye contact, smile, and maintain a reassuring tone of voice (Taylor et al, 2019)
- Engage in perspective taking: Understand stakeholder perspective and respond with empathy and compassion by relating to lived experiences (Taylor et al, 2019)

Trauma Informed Care (2.15, 2.19, 3.01)

- Acknowledge trauma and its potential impact: Avoid retraumatization by understanding that features of the environment may share similar properties of a past traumatic event. (Rajaraman et al, 2022)
- Ensure safety and trust: Promote during first interactions with clients and continue for the duration of care. Ensure emotional and physical safety and being free from aversive stimulation (Rajaraman et al, 2022)
- Promote choice and shared governance: Ensure client voice in therapeutic process, emphasize on skill development, prioritize client assent (Rajaraman et al, 2022)

Culturally Responsive Care (1.07, 1.10, 2.09, 2.13, 2.14, 3.01)

- Intake: focus on collecting and using relevant demographic information, building rapport with caregivers, and selecting goals in a culturally responsive manner through perspective taking and cultural humility. To facilitate rapport building, practitioners should collaborate, maintain positive social interactions, and build skills related to empathy and compassion (Jimenez-Gomez & Beaulieu, 2022)
- Functional Analysis: Gather information on specific conditions that evoke problem behavior and specific consequences that follow. Consider creating test conditions that incorporate cultural variables to simulate clients' natural environment. (Jimenez-Gomez & Beaulieu)
- Intervention: awareness of relevant cultural variables and how they impact goal selection (such as meals or toileting). Capture the relevant aspects of behavior in operational definitions to make a socially significant change (culturally appropriate social initiation behaviors) (Jimenez-Gomez & Beaulieu, 2022)
- Unproven therapies: Provide resources to evidence-based treatment; however, be open to evaluating unproven treatments based on caregiver preferences. Consider evaluating with single-subject design. (Jimenez-Gomez & Beaulieu, 2022)

Toward an Efficient & Effective Training of Preference Assessments: A Flow Chart and Asynchronous BST Module for MSWO

Viridiana Ruiz, Evelyn Brito, Debra Berry Malmberg, Chunying Jin, Nelly Terteryan

Introduction

Purpose

- This study aims to systematically replicate and expand Higgins et al. (2017) by evaluating the efficacy of flow charts and asynchronous behavior skills training for conducting an MSWO preference assessment.
- The importance of this study is to develop a preference assessment training method that is efficient and effective for staff member to use.

Literature Review

Figure 1. Preference assessment training methods found in JABA from 2000-2023

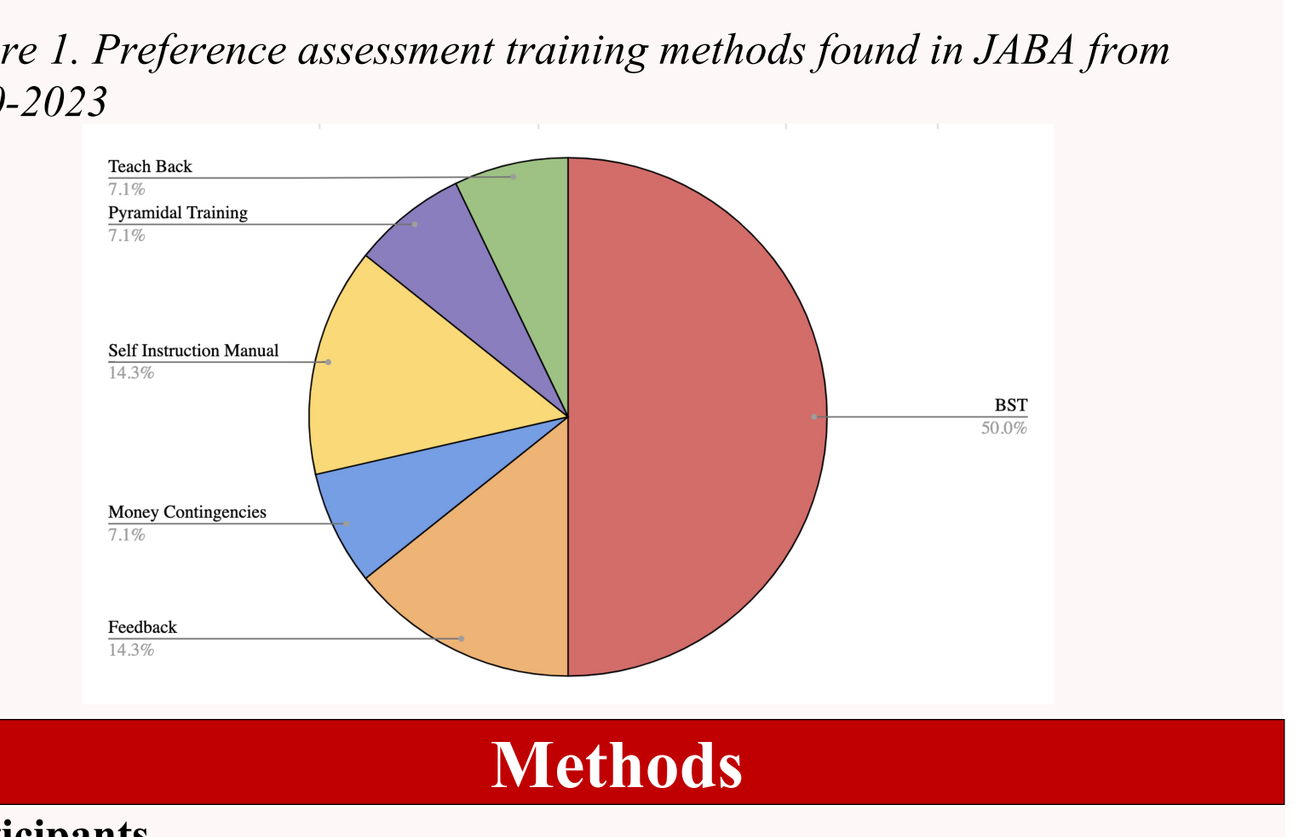
Teach Back
Pyramidal Training
Self Instruction Manual
Money Contingencies
Feedback
BST

Methods

Participants
- We aim to include 12 college students at a four-year state institution

Setting
- University-based Autism Clinic
 - Therapy rooms will contain a table, a chair, and MSWO stimuli

Dependent Variable(s)
- 14 skill components see Table 1.

Data Collection
- Trial-based observation
- Paper and pencil

Independent Variable(s)
- Flow chart: A visual aid detailing the 14 component skills that facilitates problem solving
- Asynchronous BST module: A module that includes instruction, video modeling, roleplay, and simulated feedback for conducting an MSWO preference assessment

Procedures
- Baseline: Instructional sheet on MSWO is provided to the participants
- Flow Chart (Phase 1): After baseline, participants who did not meet mastery criteria of 90% are provided with the flow chart
- Asynchronous BST Module (Phase 2): Participants who did not meet the mastery criteria of 90% in the previous flow chart phase are provided with an asynchronous BST module
- In-vivo BST Training (Phase 3): After the flow chart and asynchronous phases, participants who did not meet the mastery criteria of 90% across 3 consecutive sessions are provided with in-vivo training/feedback until mastery criteria is met
- Generalization: Probes were conducted across one novel person and one novel item

Experimental Design
- Multiple baseline across participants

Data and Graphical Analysis

Dependent Variables	Operational Defenitions
Standard Skills	
1. Pre-session exposure	Before the first trial (of the first assessment only), the observer scored a correct response if the participant presented each item to the child for 30 s (+/−2).
2. Item arrangement & prompt	For each trial, the observer scored a correct response if the participant placed all items in a straight line or arc in front of the child and instructed the child to "pick one" while directly looking at the client and not the array of stimuli
3. Record selections	For each trial, the observer scored a correct response if the participant recorded child selections on each trial and a second correct response was scored if the trainee accurately summarized the child data.
4. Rotate items	For each trial, the observer scored a correct response if the participant correctly rotated the remaining items.
5. Calculate & rank items	For each trial, the observer scored a correct response if the participant accurately summarized the child data.
Skills for an appropriate item selection	
6. Item provided	If the child selected an item within 30 s, the observer scored a correct response if the participant provided access to that item for 30 s.
7. Item removed	After 30 s, the observer scored a correct response if the participant removed the selected item.
Skills for a sequential item selection	
8. 1st selected item is provided	If the child selected two items sequentially, the observer scored a correct response if the participant provided access to the item selected first for 30 s
9. Item removed	After 30 s, the observer scored a correct response if the participant removed the selected item.
Skills for a simultaneous item selection	
10. Block access & rotate items	If the child selected two items simultaneously, the observer scored a correct response if the participant blocked access to the items and reinitiated the trial.
11. End session & remove items	If the child simultaneously selects the items again, the observer scored a correct response if the participant removed all items and ended the session.
Skill for the selection of an item not in the array	
12. Block access	If the child grabbed another item while he or she had access to the one selected, the observer scored a correct response if the participant blocked access to the item and continued with the current trial.
Skill for when no item is selected	
13. End session & remove items	If the child did not select an item within 30 s, the observer scored a correct response if the participant removed all items and terminated the current assessment.
Skill for problem behavior	
14. End session & remove items	If the child engages in problem behavior, the observer scored a correct response if the participant removed all items and terminated the current assessment.

Table 1. Depicts Higgins et al. 13 components with modifications and additions.

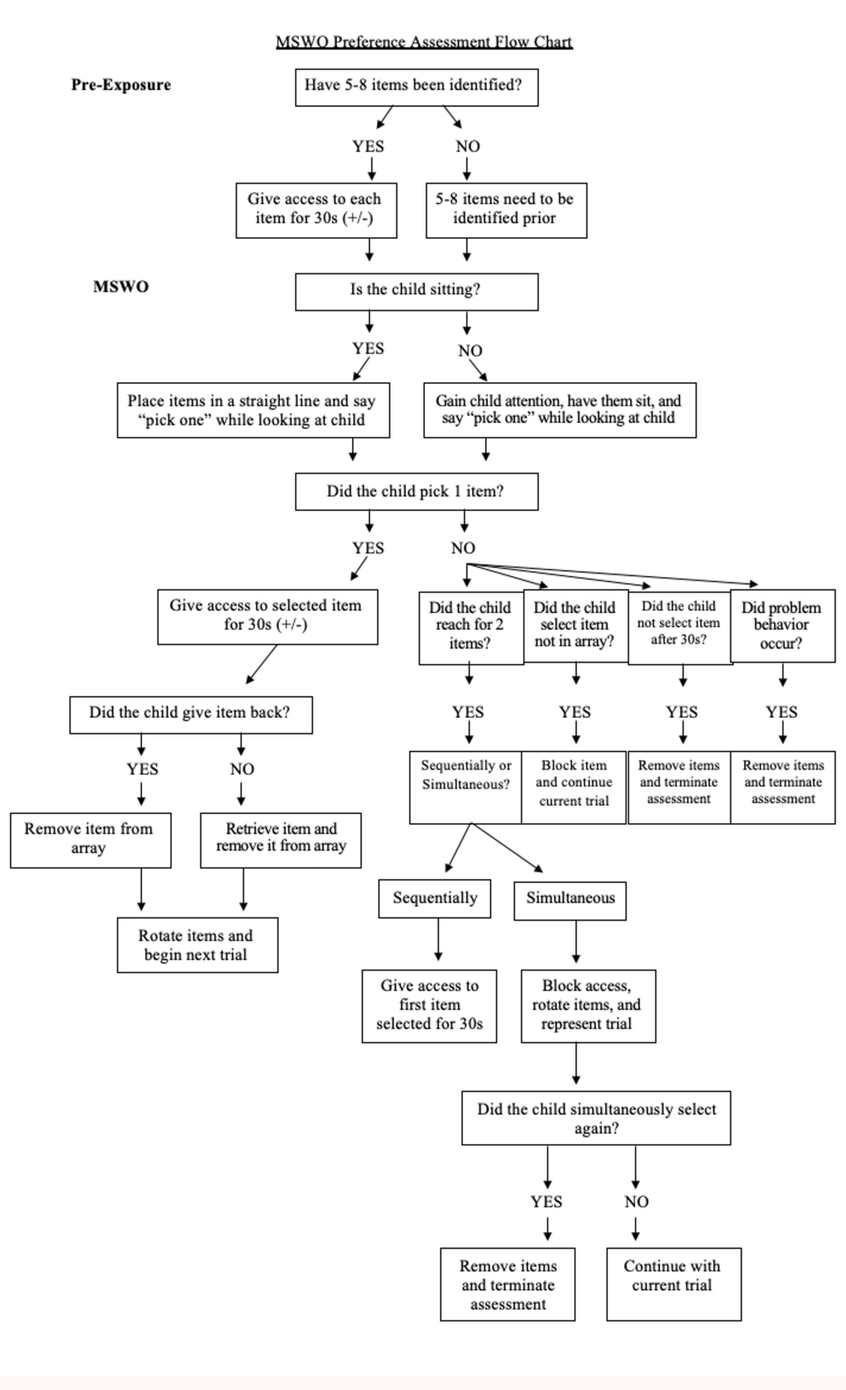
MSWO Preference Assessment Flow Chart
Pre-Exposure
Have 5-8 items been identified?
YES
NO
Give access to each item for 30s (+/-)
5-8 items need to be identified prior
MSWO
Is the child sitting?
Place items in a straight line and say "pick one" while looking at child
Gain child attention, have them sit, and say "pick one" while looking at child
Did the child pick 1 item?
Give access to selected item for 30s (+/-)
Did the child reach for 2 items?
Did the child select item not in array?
Did the child not select item after 30s?
Did problem behavior occur?

Phase 1: Flow Chart

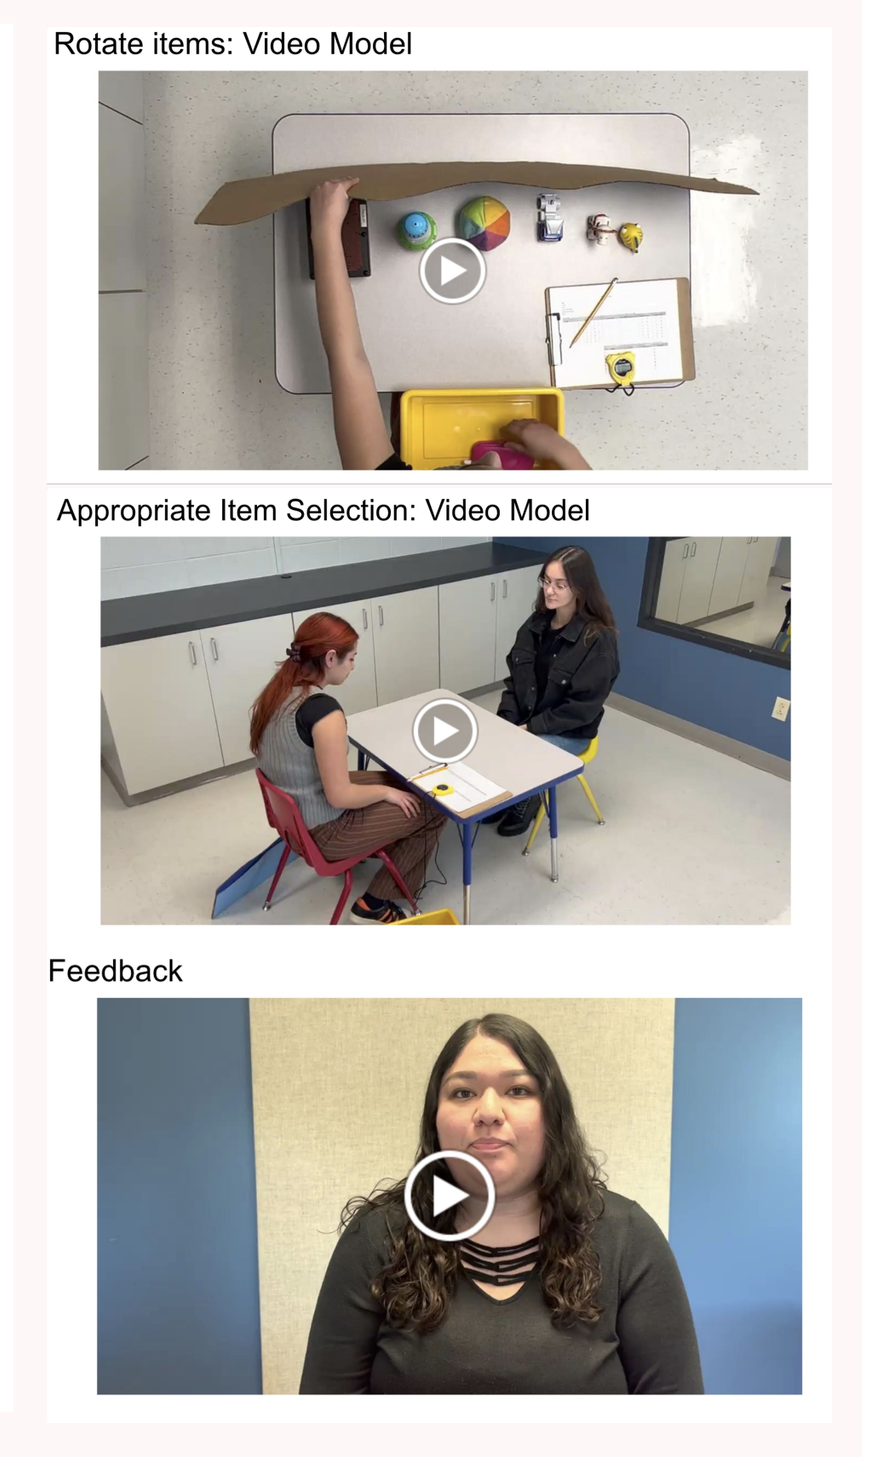
Rotate items: Video Model
Appropriate Item Selection: Video Model
Feedback

Phase 2: Asynchronous BST Module

Results

- We anticipate the following results - see Figure 2.
 - Participants' performance to improve but not meet the mastery criteria upon introduction of the flow chart
 - Majority of participants' performance to reach mastery criteria upon introduction of asynchronous BST module
 - Small number of participants' performance to need in-vivo BST training
 - Rate of mastery criteria of 90% across 3 consecutive sessions being dependent on the skill repertoire of each participant

Hypothetical Data

Figure 2. Hypothetical Data

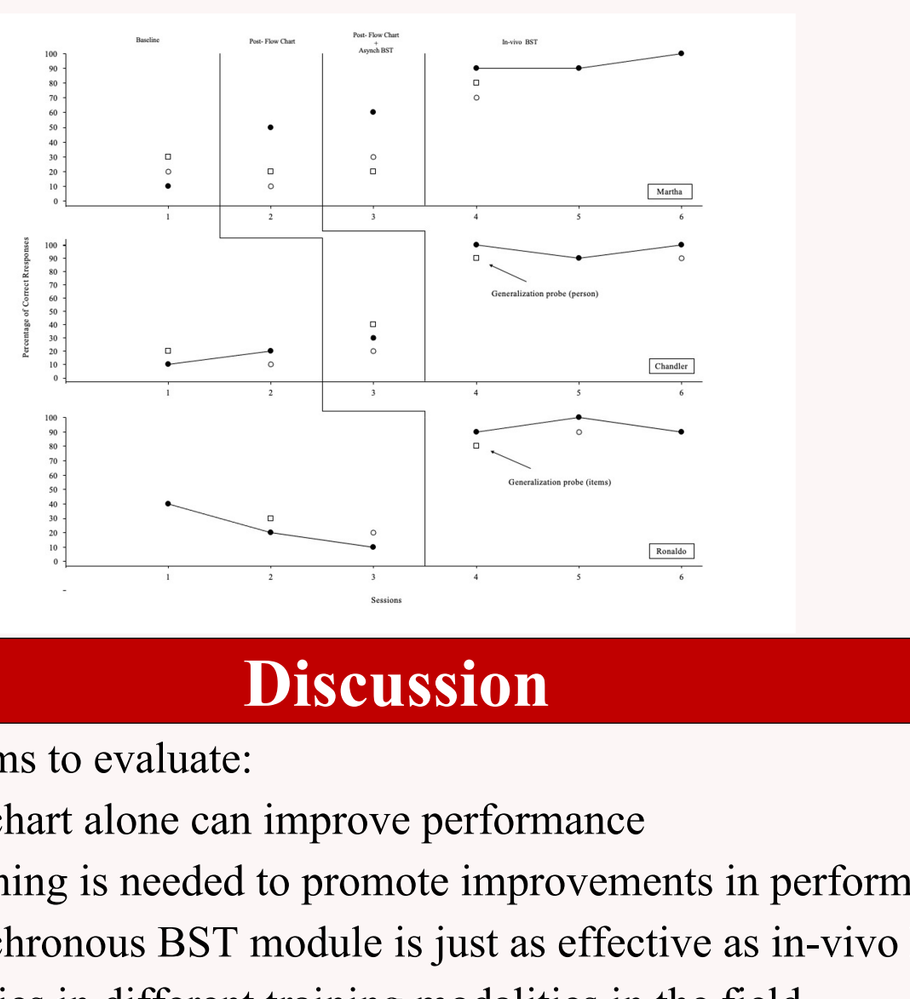

Discussion

- This study aims to evaluate:
 - If the flowchart alone can improve performance
 - If BST training is needed to promote improvements in performance
 - If the asynchronous BST module is just as effective as in-vivo BST
 - Discrepancies in different training modalities in the field.
- This study improves the cost effectiveness and MSWO training efficiency
- Future research could include efficient training strategies for other preference assessments and assessing the present training package with a client in a clinical context

References

DeLeon, I. G., & Iwata, B. A. (1996). Evaluation of a multiple-stimulus presentation format for assessing reinforcer preferences. Journal of Applied Behavior Analysis, 29(4), 519–533. https://doi.org/10.1901/jaba.1996.29-519
Graff, R. B., & Karsten, A. M. (2012). Evaluation of a self-instruction package for conducting stimulus preference assessments. Journal of Applied Behavior Analysis, 45(1), 69–82. https://doi.org/10.1901/jaba.2012.45-69
Higgins, W. J., Luczynski, K. C., Carroll, R. A., Fisher, W. W., & Mudford, O. C. (2017). Evaluation of a telehealth training package to remotely train staff to conduct a preference assessment. Journal of Applied Behavior Analysis, 50(2), 238–251. https://doi.org/10.1002/jaba.370
Ramon, D., Yu, C. T., Martin, G. L., & Martin, T. (2015). Evaluation of a self-instructional manual to teach multiple-stimulus without replacement preference assessments. Journal of Behavioral Education, 24(3), 289–303. https://doi.org/10.1007/s10864-015-9222-3
Roscoe, E. M., & Fisher, W. W. (2008). Evaluation of an efficient method for training staff to implement stimulus preference assessments. Journal of Applied Behavior Analysis, 41(2), 249–254. https://doi.org/10.1901/jaba.2008.41-249
Weldy, C. R., Rapp, J. T., & Capocasa, K. (2014). Training staff to implement brief stimulus preference assessments. Journal of Applied Behavior Analysis, 47(1), 214–218. https://doi.org/10.1002/jaba.98

Review of Compassionate Care in Supervisory Relationships in Applied Behavior Analysis

Lizzette Graciano and Ya-Chih Chang
California State University, Los Angeles

Introduction

- Therapeutic fields, such as psychology and mental health counseling, train their professionals in therapeutic relationships and compassionate care (e.g., Melton et al., 2023). However, many BCBAs in supervisory positions claimed that they had not received formal training in compassionate practices or relationship-building techniques for supervision (LeBlanc et al. 2020).
- Aside from the context related to family satisfaction and treatment adherence (e.g., LeBlanc et al., 2019; Taylor et al., 2019), previous studies on compassionate practice and therapeutic skills have not examined its positive impact in other appropriate contexts, such as supervisor and supervisee relationships.
- A specific focus given to training prospective BCBAs in compassionate skills may promote positive supervisor and supervisee relationships.

Specific Aim

- This presentation will review the studies that examine the role of compassion and its impact on applied behavior analysis (ABA) supervisor and supervisee relationships.

Methods

The literature review was initiated in March 2023. One database (PsychInfo) was used for the literature search. Keyword search terms used for the review included empathy, compassion, compassionate care, compassionate practices, training, supervision, therapeutic skills, soft skills and ABA.

Only studies that examined the relationship of supervisors and supervisee relationships were included in the study. Four studies were included in the review.

Definition of Compassionate Practices

Compassionate Practices (Taylor et al., 2019)
- Respect of the cultural values and beliefs of staff.
- Using language appropriate to the staff's skill level
- Demonstrating patience while training
- Reassure staff when they question their abilities
- Ask staff how they are doing
- Asking for staff input
- Acknowledges their own mistakes
- Establishes clear expectations
- Acknowledges staff feeling when discussing difficult feedback
- Being available to meet staff.
- Asking staff how they are doing.

Attributes (Melton et al., 2023)
- Identify information about someone's contingencies.
- Understand types of suffering states.
- Expression of confirming changing contingencies.
- Engage in helping action directed towards the individuals need (e.g., verbal statements of future actions, physical interaction, adding stimuli that are directly related to the suffering state, or removing stimuli that is observed to cause suffering).

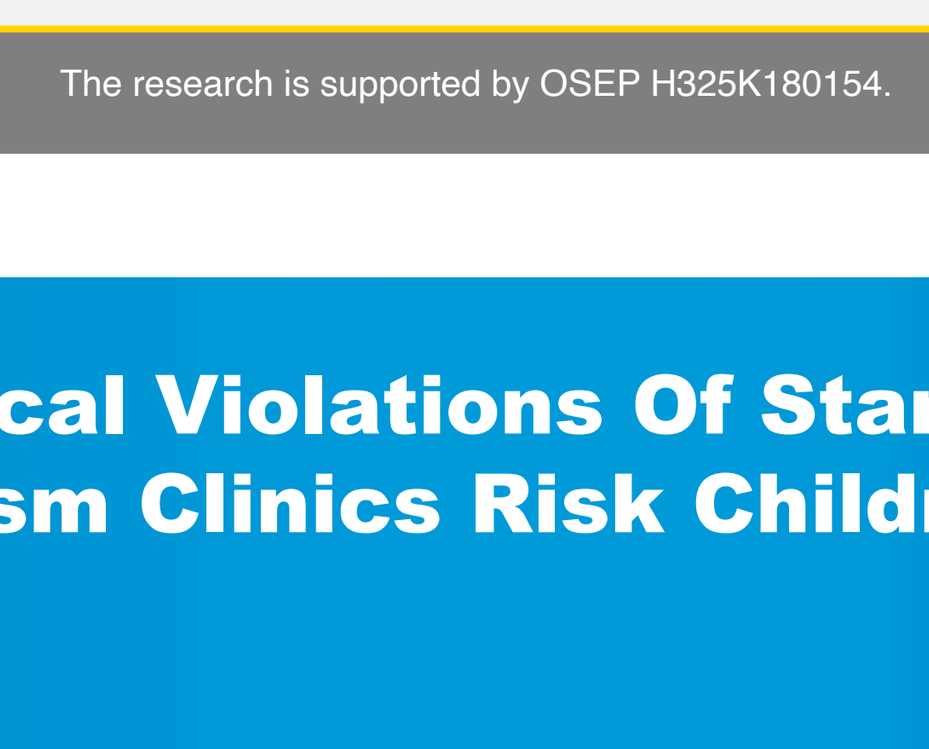

The research is supported by OSEP H325K180154.

Results

Compassionate care has been widely studied in the therapeutic field, including the field of ABA. However, most of the studies have examined compassionate care between therapists and clients. Even less studies have measured and examined the effects of compassionate care between supervisors and supervisees (e.g., Hess et al., 2022; Taylor et al., 2019). Three themes were identified in the review:

1) Definition of compassionate care varied across the studies.
2) One study used therapeutic skills training to help supervisors build relationships with their supervisees (Hess et al., 2022).
3) Only two studies included outcomes of supervisory skills, including compassionate care (Hess et al., 2022; Taylor et al., 2019).

Implications

The preliminary findings from the review suggests positive effects of compassionate care between supervisors and supervisee in ABA. Future studies should measure the direct impact these practices have between supervisors and supervisees, including the indirect effects it may have on supervisee's clients.

References

Hess, A., Garcia, Y., Gould, E., & Catagnus, R. (2022). Enhancing supervisory relationships with values and committed action training. *Journal of Contextual Behavior Science, 26*(2022) 241-252.

Melton, B., O'Connell-Sussman, E., Lord, J., & Weiss, M.J. (2023) Empathy and compassion as the radical behaviorist views it: A conceptual analysis. *Behavior Analysis in Practice,*

Rohrer, J. L., Marshall, K.B., Suzio, C., & Weiss, M.J. (2021). Soft skills: The case for compassionate approaches or how behavior analysis keeps finding its heart. *Behavior Analysis in Practice, 14* (4), 1135-1143.

Taylor, B.A., LeBlanc, L.A., & Nosik, M.R. (2019). Compassionate care in behavior analytic treatment : can outcomes be enhanced by attending to relationships with caregivers? *Behavior Analysis in Practice, 12*(3), 654-666.

Social Media Publications: Ethical Violations Of Stand-Alone Applied Behavior Analysis Autism Clinics Risk Children's Safety

Alexandra Ingebritson
Center for Behavior Analysis, University of West Florida

Introduction

Updates to the 2022 Behavior Analysis Certification Board (BACB) Ethics Code specified which social media content is permitted for publication by behavior analysts. To protect the privacy and safety of their clients, these updates in code 5.10 now require that for each publication that includes images of their clients' faces, informed consent is obtained before publishing, and these publications include a disclaimer stating informed consent was obtained (Behavior Analyst Certification Board, 2020, p. 17).

Purpose

The aim of this study was to review professional social media channels for stand-alone Applied Behavior Analysis (ABA) clinics that treat children with autism spectrum disorder (ASD) who share images of their clients. The goal in this analysis was to determine the overall level of compliance with the BACB's updated codes.

Methodology

Using a convenience sampling for social media content analysis method (Kim et al., 2018), an internet search was conducted to investigate how behavior analysts are sharing images of their clients on social media. To conduct this search, terms such as "ABA Clinic," "Applied Behavior Analysis Center," and "ABA Therapy" were used. To avoid a geographical location bias, GPS location services were turned off on devices used to compile the dataset.

Once analyzed, each qualifying social media channel was sorted into one of two groups: "Group 1: Compliant with the 2022 BACB Ethics Code" or "Group 2: Violators of the 2022 BACB Ethics Code."

From September through December of 2022, public Facebook social media channels of 92 different ABA clinics, which meet inclusion criteria, were analyzed.

Figure 1
Examples of social media post sorted into group 1 (left) and social media posts sorted into group 2 (right)

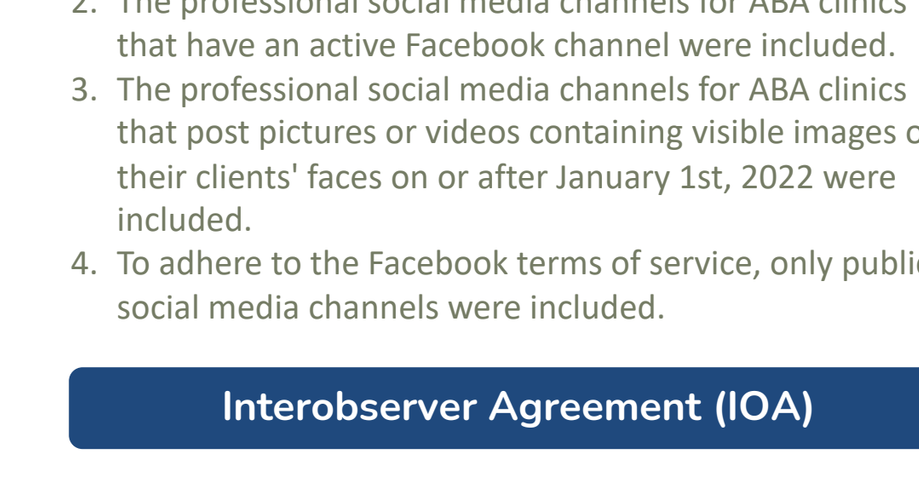
Sample ABA Clinic
Sample ABA Clinic

Inclusion Criteria

The professional social media channels who met the following inclusion criteria were included in the dataset:

1. Stand-alone ABA clinics that employ behavior analysts who offer services for individuals with autism spectrum disorder were considered.
2. The professional social media channels for ABA clinics that have an active Facebook channel were included.
3. The professional social media channels for ABA clinics that post pictures or videos containing visible images of their clients' faces on or after January 1st, 2022 were included.
4. To adhere to the Facebook terms of service, only public social media channels were included.

Interobserver Agreement (IOA)

The trial-by-trial IOA method (Cooper et al., 2019, pg. 113) was used to ensure validity of the data analysis. Each social media channel that was included in the dataset was treated as individual trials when they were independently analyzed by the principal investigator and the coinvestigator.

IOA Results: 100% agreement on the dataset between the principal investigator and the coinvestigator

Results

92 professional social media channels of stand-alone Applied Behavior Analysis clinics met inclusion criteria. Of the 92 social media channels that were analyzed, 13 of them were sorted into group 1, and 79 were sorted into group 2.

14.13% of behavior analysts' professional social media channels included in the dataset complied with the BACB ethics code. This means that clients who are posted on 85.87% of behavior analysts' professional social media channels included in our dataset are not protected.

Figure 2
Social Media Analysis Results

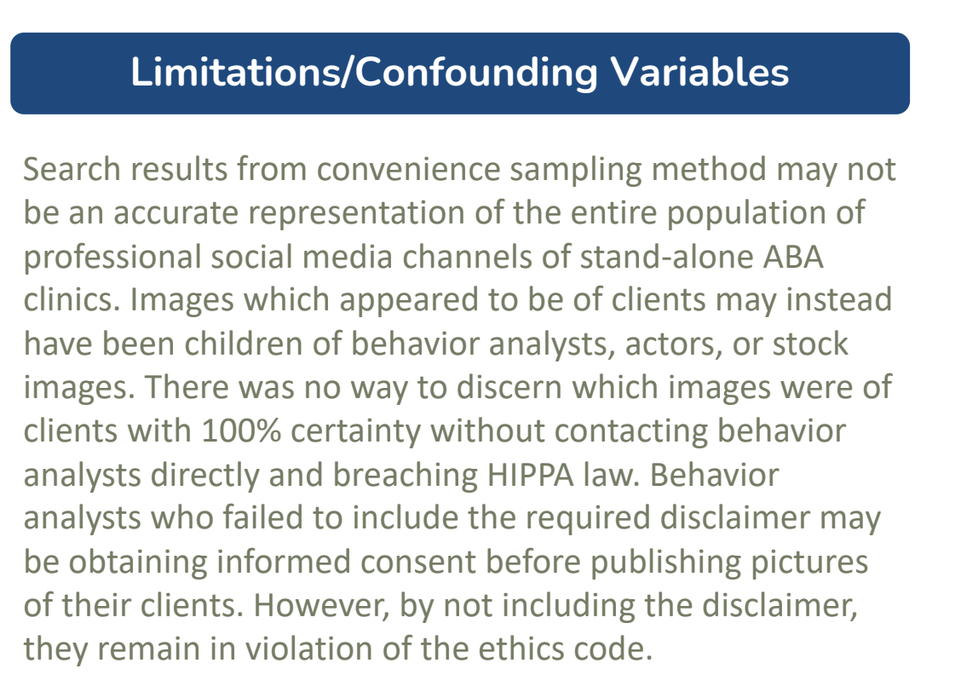
Stand-Alone Applied Behavior Analysis Autism Clinics Compliant with BACB Ethics Code 5.10
Percent of Total Clinics
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
14.13%
85.87%
Compliant
Noncompliant
Applied Behavior Analysis Autism Clinics

Limitations/Confounding Variables

Search results from convenience sampling method may not be an accurate representation of the entire population of professional social media channels of stand-alone ABA clinics. Images which appeared to be of clients may instead have been children of behavior analysts, actors, or stock images. There was no way to discern which images were of clients with 100% certainty without contacting behavior analysts directly and breaching HIPPA law. Behavior analysts who failed to include the required disclaimer may be obtaining informed consent before publishing pictures of their clients. However, by not including the disclaimer, they remain in violation of the ethics code.

Conclusion/Discussion

If behavior analysts find evidence of ethical violations in social media publications from their company, it is recommended that they notify their employers and work together with them to remedy the violations and integrate policies into their practices on gaining informed consent before posting images of their clients.

Posting images of clients on social media can unintentionally expose these children to serious risks, including sexual exploitation, future emotional distress, and even digital kidnapping (Ferrara et al., 2023). We urge behavior analysts to think twice before sharing images of their clients in any way, especially through social media. These children are already a vulnerable population and need as much of our help and protection as possible. By doing this, we can advocate for our clients!

References

Behavior Analyst Certification Board. (2020). *Ethics code for behavior analysts*. https://bacb.com/wp-content/ethics-code-for-behavior-analysts/

Cooper, J. O., Heron, T. E., & Heward, W. L. (2019). *Applied Behavior Analysis (3rd Edition)*. Hoboken, NJ: Pearson Education.

Ferrara P, Cammisa L, Corsello G, Giardino I, Vural M, Pop TL, Pettoello Mantovani C, Indrio F, Pettoello-Mantovani M, Online "Sharenting": The Dangers of Posting Sensitive Information About Children on Social Media, *The Journal of Pediatrics* (2023), doi: https://doi.org/10.1016/j.jpeds.2023.01.002.

Kim, H., Jang, S. M., Kim, S.-H., & Wan, A. (2018). *Evaluating Sampling Methods for Content Analysis of Twitter Data. Social Media + Society*, 4(2). https://doi.org/10.1177/2056305118772836

Acknowledgements

University of West Florida Faculty Advisor: Sarah Kent
Supervising BCBA: Jennifer Andoscia
Co-Investigator: Lauren Hanley

uwf.edu/ABA